This piece of material is only intended for UK Healthcare Professionals to view online; it is not intended to be printed.

OCREVUS®▼(ocrelizumab)

Ocrevus is indicated for the treatment of adult patients with relapsing forms of multiple sclerosis (RMS) with active disease defined by clinical or imaging features.
Ocrevus is indicated for the treatment of adult patients with early primary progressive multiple sclerosis (PPMS) in terms of disease duration and level of disability, and with imaging features characteristic of inflammatory activity.

Click here to access the Prescribing Information for Great Britain

Click here to access the Prescribing Information for Northern Ireland

▼ This medicinal product is subject to additional monitoring. This will allow quick identification of new safety information. Healthcare professionals are asked to report any suspected adverse reactions. Adverse events should be reported. Reporting forms and information can be found at www.mhra.gov.uk/yellowcard or search for MHRA Yellow Card in the Google Play or Apple App Store. Adverse events should also be reported to Roche Products Ltd. Please contact Roche Drug Safety Centre by emailing welwyn.uk_dsc@roche.com or calling +44 (0)1707 367554. As OCREVUS is a biological medicine, healthcare professionals should report adverse reactions by brand name and batch number.

M-GB-00003250 April 2021

Please note: QR codes from the poster have been removed

Cases Reported as Progressive Multifocal Leukoencephalopathy in Ocrelizumab-Treated Patients With Multiple Sclerosis

DB Clifford,[1] A Gass,[2] N Richert,[3] C Tornatore,[4] P Vermersch,[5] R Hughes,[6] H Koendgen,[6] R Gold[7]

[1]Washington University School of Medicine, St. Louis, MO, USA; [2]University Hospital Mannheim, Heidelberg, Germany; [3]NeuroRx Research, Montreal, QC, Canada; [4]Georgetown University Hospital, Washington DC, USA; [5]University of Lille, Lille, France; [6]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [7]Ruhr-University Bochum, Faculty of Medicine, St. Josef-Hospital, Bochum, Germany

CONCLUSIONS

- No unconfounded PML cases associated with ocrelizumab therapy have been reported
- The confirmed cases of PML in ocrelizumab-treated patients have been associated with and confounded by prior MS treatment (natalizumab, fingolimod), usually referred to as cases of carry-over PML
 - — None of these cases resulted in fatality at the time of last follow-up
 - — Approximately 17% (19,000) of patients switched from natalizumab to ocrelizumab globally. It is possible that many of these patients did so due to a higher individual risk of PML with continued natalizumab therapy
 - — The observed number of carry-over PML cases in patients switching from natalizumab to ocrelizumab is low
- Updates on safety information relating to cases reported as PML in ocrelizumab-treated patients are available at: www.ocrelizumabinfo.global and www.ocrelizumabinfo.com
- Physicians must remain vigilant for signs and symptoms of PML (clinical, MRI) when switching from MS therapies associated with PML to a new MS therapy
- If PML is suspected, dosing with ocrelizumab must be withheld
- The benefit/risk of ocrelizumab for the treatment of MS remains unchanged

INTRODUCTION AND PURPOSE

- Ocrelizumab (OCR), a humanised monoclonal antibody that targets and selectively depletes CD20+ B cells, is approved for the treatment of relapsing and primary progressive multiple sclerosis
- Progressive multifocal leukoencephalopathy (PML) is an opportunistic viral infection of the brain caused by the JC virus, typically occurring in immunocompromised patients, and may result in death or severe disability[1]
 - — JC virus is prevalent in ≥60% of the adult population as a latent or persistent infection
- The vast majority of cases of PML in patients with multiple sclerosis (MS) have been associated with and described in those treated with natalizumab[2]
- Cases of PML in patients with MS have been associated with other disease-modifying therapies (DMTs), e.g. fingolimod and dimethyl fumarate
- PML cases were also reported with anti-CD20 therapies in a variety of different disease areas and/or concomitant immunosuppressive therapies
- The purpose of these analyses was to describe cases reported as PML in OCR-treated patients with MS

METHODS

Case Report Information

- For all cases, follow-up with the treating physician was attempted at several time points
- Individual patient case narratives and brain MRI images prior to, and after, the initiation of OCR therapy (where available) were reviewed up to 31 July 2019
- All assessments were supported by an external panel of expert advisors
- Descriptive statistics were used

PML Diagnostic Criteria

- The certainty of PML diagnosis was graded per American Association of Neurology (AAN) diagnostic criteria and considered "confirmed" if all criteria for the classification of "definite" were met (see **Table 1**)[3]
- Definition of carry-over PML: PML that develops a few months after stopping a DMT associated with PML and starting a different DMT[4]
 - — In these cases, PML could have developed without causing symptoms while the patient was still on the previous DMT, or shortly after stopping the previous DMT
- Post-marketing exposure estimations in the USA were based on Symphony Health claims and distributor shipment data; European Economic Area and Rest of World estimations are based on total number of OCR vials sold (monthly basis) and IQVIA LRx (Germany) data

Table 1. AAN diagnostic criteria for PML[3]

Certainty of PML diagnosis	Compatible clinical features	Compatible imaging findings	CSF PCR for JC virus
Definite	+	+	+
Probable	+ –	– +	+ +
Possible	+ –	+ –	– / ND +
Not PML	– + –	– – +	– – –

CSF, cerebrospinal fluid; ND, not determined; PCR, polymerase chain reaction; PML, progressive multifocal leukoencephalopathy

Figure 1. Summary of confirmed carry-over PML cases

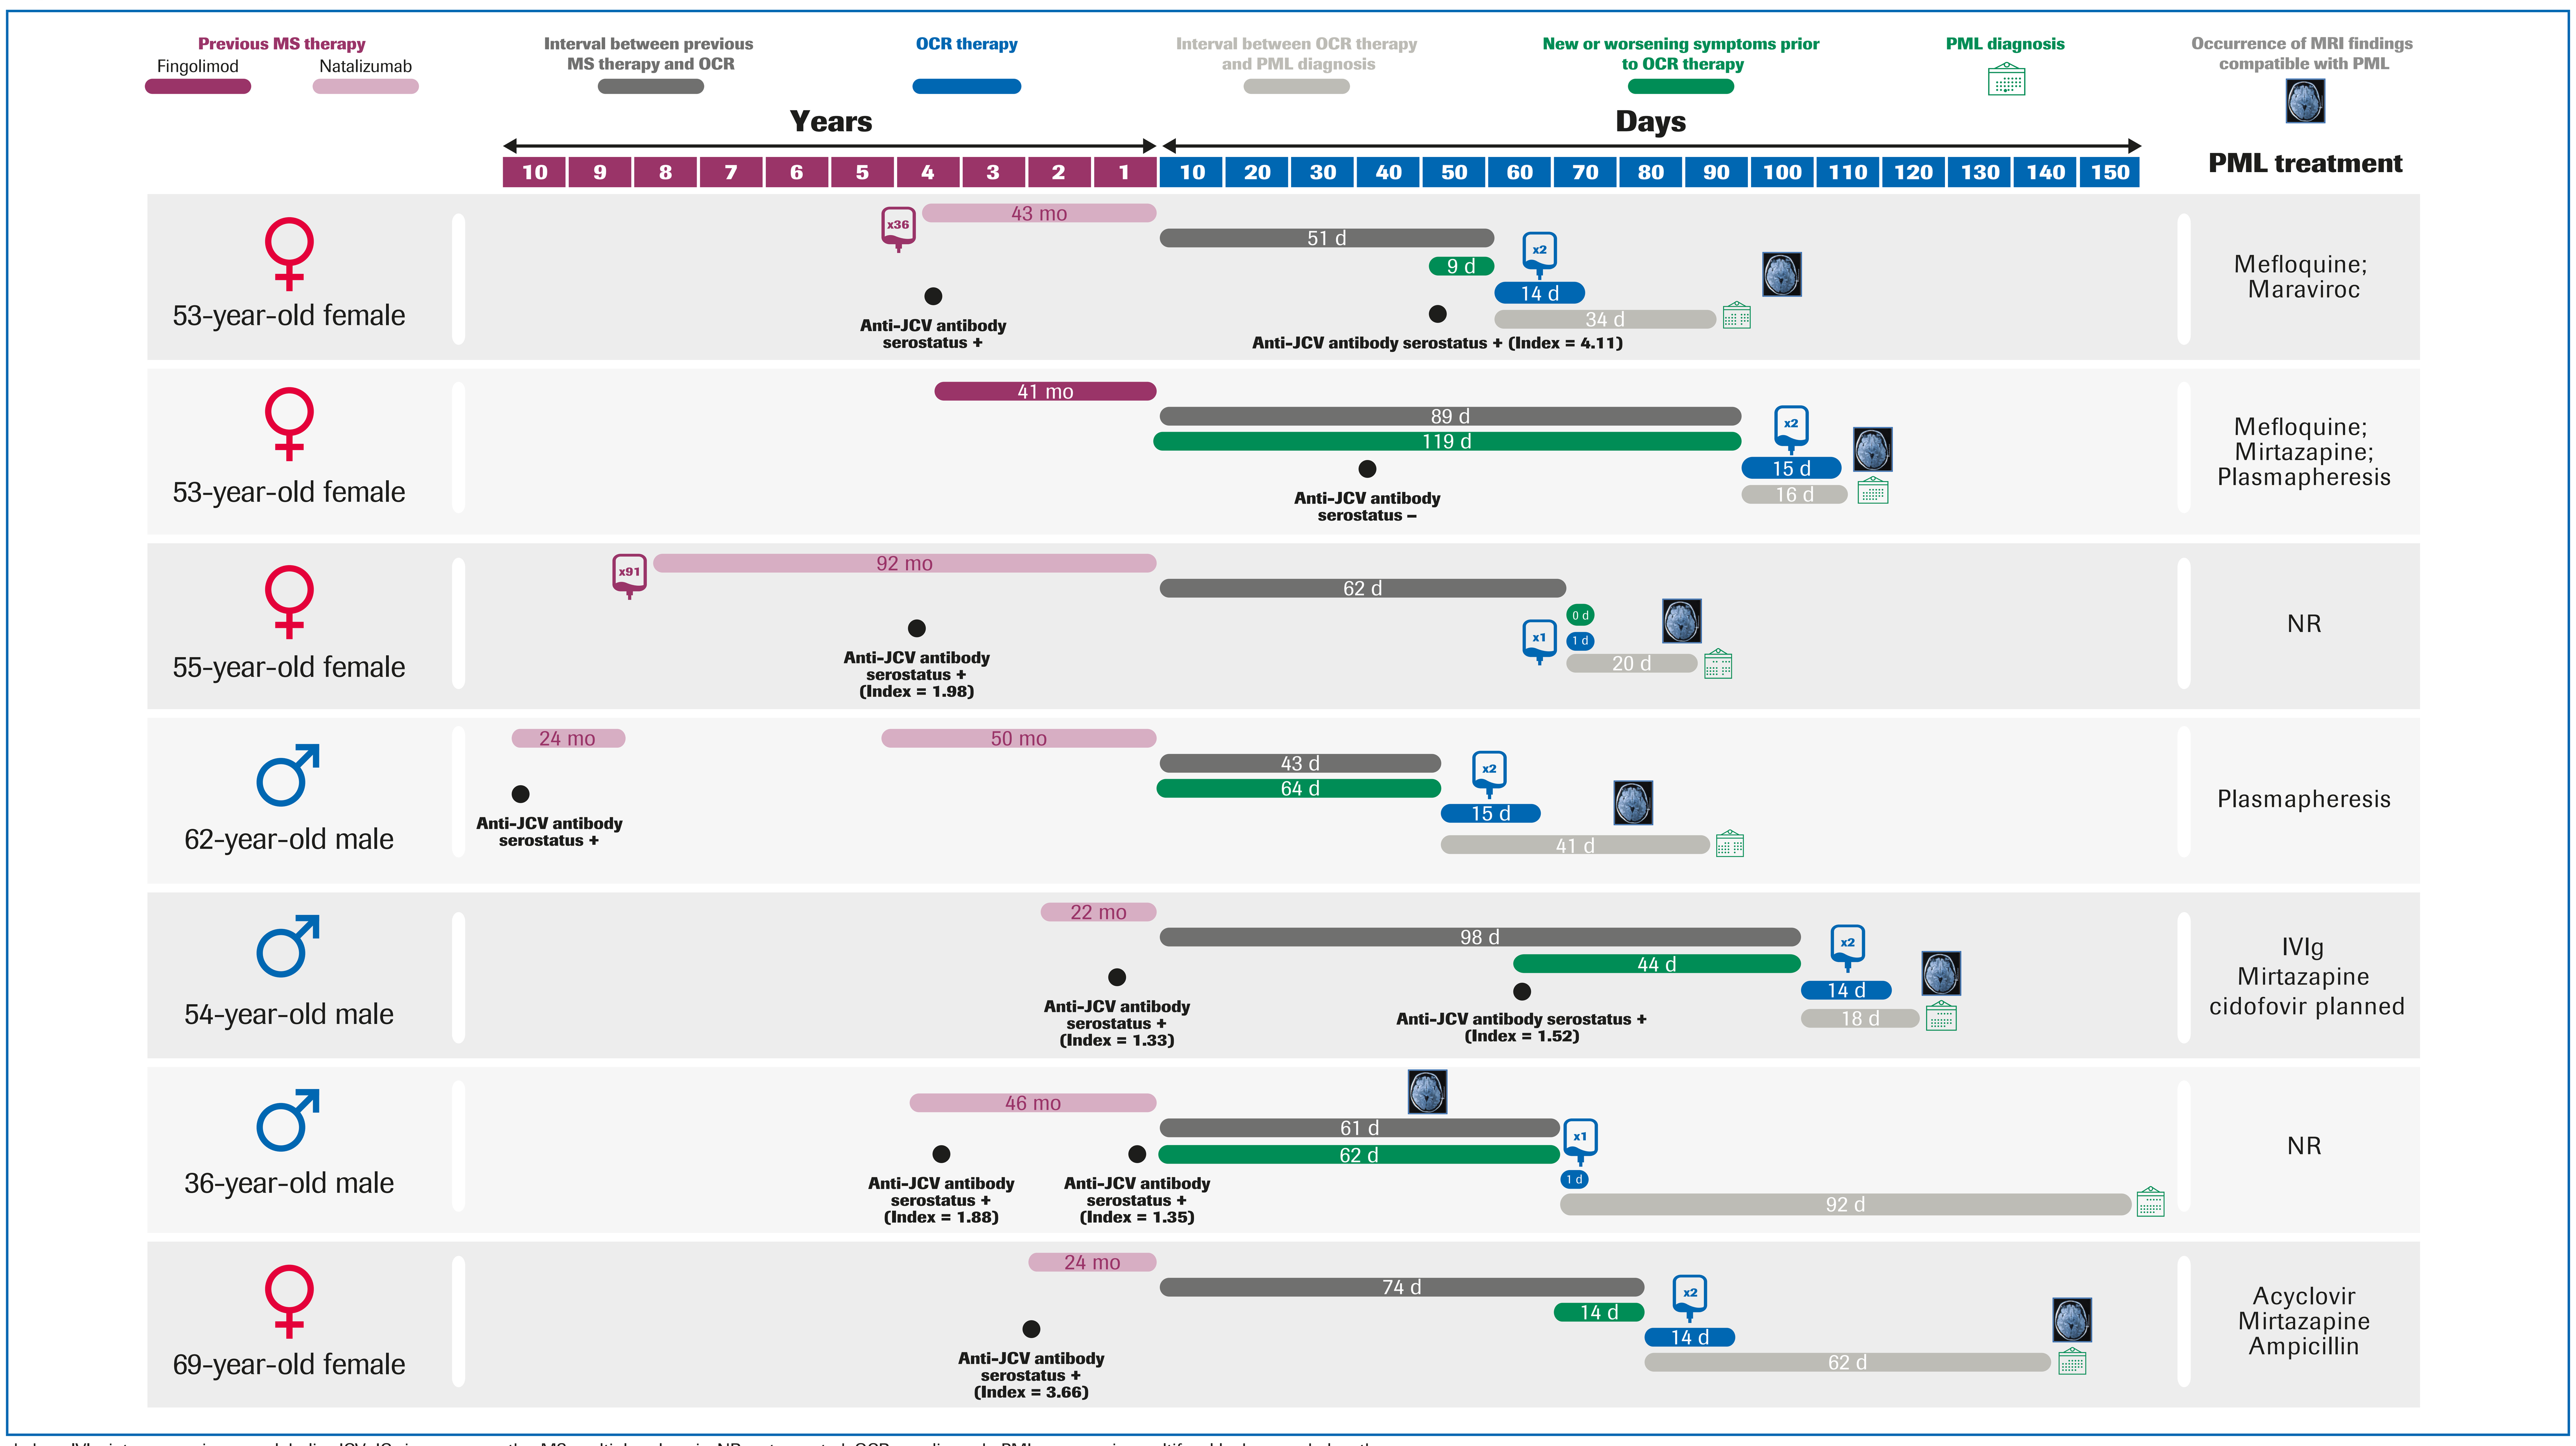
Previous MS therapy
Fingolimod
Natalizumab
Interval between previous MS therapy and OCR
OCR therapy
Interval between OCR therapy and PML diagnosis
New or worsening symptoms prior to OCR therapy
PML diagnosis
Occurrence of MRI findings compatible with PML
Years
Days
PML treatment
53-year-old female
Anti-JCV antibody serostatus +
Anti-JCV antibody serostatus + (Index = 4.11)
Mefloquine; Maraviroc
53-year-old female
Anti-JCV antibody serostatus –
Mefloquine; Mirtazapine; Plasmapheresis
55-year-old female
Anti-JCV antibody serostatus + (Index = 1.98)
NR
62-year-old male
Anti-JCV antibody serostatus +
Plasmapheresis
54-year-old male
Anti-JCV antibody serostatus + (Index = 1.33)
Anti-JCV antibody serostatus + (Index = 1.52)
IVIg Mirtazapine cidofovir planned
36-year-old male
Anti-JCV antibody serostatus + (Index = 1.88)
Anti-JCV antibody serostatus + (Index = 1.35)
NR
69-year-old female
Anti-JCV antibody serostatus + (Index = 3.66)
Acyclovir Mirtazapine Ampicillin

d, days; IVIg, intravenous immunoglobulin; JCV, JC virus; mo, months; MS, multiple sclerosis; NR, not reported; OCR, ocrelizumab; PML, progressive multifocal leukoencephalopathy.

RESULTS

Global OCR Patient Exposure

- As of 31 July 2019, more than 120,000 patients with MS have been exposed to OCR globally
 - — Clinical trials: more than 6,000 patients
 - — Post-marketing experience: more than 114,000 patients
 - — Approximately 17% (19,000) of post-marketing patients treated with OCR globally have previously received natalizumab

Overall Summary of Cases Reported as PML

- As of 31 July 2019, according to AAN diagnostic criteria for PML, there are:
 - — Seven confirmed carry-over cases of definite PML
 - — Five unconfirmed carry-over cases reported as PML, or suspicion of PML, that do not meet AAN PML diagnostic criteria for definite PML
 - — One case with insufficient information for assessment
- All 12 cases with information available were confounded by prior MS DMTs (natalizumab, n=11; fingolimod, n=1)

Confirmed PML Cases (All Carry-Over)

- The seven confirmed carry-over PML cases are summarised in **Figure 1**
 - — Six patients had previously received treatment with natalizumab (treatment duration, 22–92 months) and all tested positive for serum anti-JC virus antibodies (index range, 1.33–4.11) prior to the diagnosis of PML
 - Time between initiation of OCR therapy and a definite diagnosis of PML: 16–92 days
 - Onset of PML in all cases was within 6 months of last dose of previous natalizumab
 - — One patient had previously received fingolimod (treatment duration: 41 months) and was anti-JCV antibody-negative prior to the diagnosis of PML
 - The time between the initiation of OCR therapy and the definite diagnosis of PML was 16 days
- All patients had new and/or worsening clinical symptoms and/or MRI findings compatible with PML prior to receiving OCR

Unconfirmed Reports of PML

- Five reports suggestive of carry-over PML but not fully meeting AAN diagnostic criteria:
 - — All five cases had MRI findings compatible with PML, one patient also had clinical symptoms and one other patient also had detection of JCV DNA in the cerebrospinal fluid
 - All reports were in patients previously treated with natalizumab and already at higher risk for a natalizumab-associated PML
- One report was an unconfirmed consumer report of a potential PML; permission for follow-up was not given by the reporter and further assessment is not possible
 - — Only age and gender were disclosed; these two details match two of the previously reported cases of carry-over PML in OCR-treated patients

Patient Outcomes

- All cases reported as PML in OCR-treated patients were non-fatal at the point of last update

ACKNOWLEDGEMENTS

Sponsored by F. Hoffmann-La Roche Ltd; writing and editorial assistance was provided by Articulate Science, UK.

REFERENCES

1. Grebenciucova E, Berger JR. *Neurol Clin* 2018;36:739–750.
2. Berger JR. *Mult Scler Relat Disord* 2017;12:59–63.
3. Berger JR, *et al. Neurology* 2013;80:1430-1438.
4. Giovannoni G, *et al. Pract Neurol* 2016;16:389–393.

DISCLOSURES

DB Clifford has received support for consulting and serving on Data Safety Monitoring Boards or Adjudication Committees for Amgen, Dr. Reddy's, Takeda, Roche, Mitsubishi Tanabe, Genentech, Shire, Biogen, Atara Biotherapeutics, Inhibikase, GSK, Wave Life Sciences, and EMD Serono; payments for legal consultations from Sal Indomenico PC and Shevlin Smith; support for a lecture by Limits, LTD; royalties from Wolters Kluwer and Up-to-Date, and is supported by NIH grants AI069439, NS10719801, MH10734504, AG0617904, AG05326702 and UL1TR00234502. A Gass has received honoraria for lecturing and advisory boards from Novartis, Biogen, Merck Serono, Sanofi-Genzyme and Roche. N Richert is an employee of NeuroRx Research. C Tornatore has received either research support, acted as a consultant or is on the speaker board of the following: Genentech, Biogen, Serono, Sanofi, and MAPI. P Vermersch has received honoraria and consulting fees from Bayer, Biogen, Celgene, MedDay, Merck, Novartis, Roche, Sanofi Genzyme, Servier, Teva and Almirall, and research support from Bayer, Biogen, Merck, Novartis, Roche and Sanofi Genzyme. R Hughes is an employee and shareholder of F. Hoffmann-La Roche Ltd. H Koendgen is an employee and shareholder of F. Hoffmann-La Roche Ltd. R Gold has received speaker's and board honoraria from Baxter, Bayer Schering, Biogen Idec, CLB Behring, Celgene, Genzyme, Janssen, Merck Serono, Novartis, Roche, Stendhal, Talecris and Teva; and is a shareholder of Merck Serono and Roche; departmental grant support has been received from Bayer Schering, BiogenIdec, Genzyme, Merck Serono, Novartis, and Teva.

Presented at the 35th Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS); 11–13 September 2019; Stockholm, Sweden

Poster P970