Design of the Ocrelizumab Pregnancy Registry to Assess Maternal, Fetal and Infant Outcomes in Women With Multiple Sclerosis Who Were Exposed to Ocrelizumab During, or Within 6 Months Before, Pregnancy

D Wormser,[1] P Engel,[2] K Hahn,[3] S Bader-Weder,[1] E-M Didden,[1] J Evershed,[4] M Garas,[1] Q Wang,[1] K Hellwig[5]

[1]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [2]IQVIA, Paris, France; [3]IQVIA, Cambridge, MA, USA; [4]Roche Products Ltd, Welwyn Garden City, UK; [5]St. Josef-Hospital, Ruhr University, Bochum, Germany

BACKGROUND

- Ocrelizumab (OCR) is a recombinant, humanized, monoclonal immunoglobulin G1 antibody that selectively targets CD20+ B cells
- Immunoglobulins such as OCR do not cross the placenta during the first trimester of pregnancy, but transfer of OCR can occur thereafter[1]
- The safety profile of OCR has been investigated in multiple clinical trials and although the use of effective contraception was mandatory, 25 pregnancies have been reported in women with multiple sclerosis (MS) receiving OCR during these trials up to the end of January 2017; in 14 of these 25 pregnancies, the fetuses were considered to have been exposed to OCR[2]
- The small number of pregnancies and pregnancy outcomes[2] that have been reported from clinical trials means the safety profile of OCR in pregnancy and fetal outcomes has yet to be established
- This study has been designed as part of the post-marketing activities to provide information that will be helpful to patients receiving OCR and clinicians when making decisions related to pregnancy
- OCR has an average terminal half-life of 26 days,[3] and based on the estimated elimination rates after the last administration, the current European and FDA label information states "Women of childbearing potential should use contraception while receiving ocrelizumab and for 12 months after the last infusion of ocrelizumab"[3] and "You should use birth control (contraception) during treatment with ocrelizumab and for 6 months after your last infusion of ocrelizumab",[4] respectively

[2]Vukusic S, *et al.* *ECTRIMS* 2017

OBJECTIVE

- To assess maternal, fetal and infant outcomes in women with MS exposed to OCR during the 6 months prior to their last menstrual period (LMP) or at any time during pregnancy

METHODS

Study Objectives

- This study will characterize pregnancy and infant outcomes of women with MS exposed to OCR during the 6 months prior to their LMP or at any time during pregnancy, including:[a]
 - The frequency of selected adverse pregnancy outcomes (e.g. spontaneous abortions, stillbirths, elective or therapeutic terminations, preterm births, C-sections, and urinary and other infections)
 - The frequency of selected adverse fetal/neonatal/infant outcomes (e.g. major congenital malformations, small for gestational age, postnatal growth and development, adverse effects on immune system development) at birth and through at least the first year of life of infants

[a]Protocol number WA40063. The final study design may be amended based on any further discussions with the regulatory authorities.

Data Sources

- Data will be obtained through questionnaires administered to patients and their healthcare professionals (HCPs; neurologist, obstetrician and pediatrician)
- This study will compare the frequency of each safety event of interest between OCR-exposed pregnant women with MS and two comparison cohorts:
 - Women with MS with no prior OCR exposure before or during pregnancy
 - Women without MS

Study Design

- The registry will collect primary data from pregnant women with MS from the United States, Germany and other potential countries, who have been exposed to OCR during the 6 months prior to their LMP or at any time during pregnancy (**Figure 1**)
 - Data will be collected from patients and their HCPs (neurologist, obstetrician) during pregnancy and through at least 1 year after birth (infant's pediatrician; **Figure 1, Table 1**)
 - The design of the pregnancy exposure registry is consistent with relevant guidelines and recommendations[5–8]
- Comparisons will be made between the observed frequency of each event (**Figure 2**) in this study with frequencies reported in other existing data sources, including, but not limited to:
 - German Multiple Sclerosis and Pregnancy Registry (Deutsches Multiple Sklerose und Kinderwunsch Register; DMSKW)
 - Metropolitan Atlanta Congenital Defects Program (MACDP)
 - European Surveillance of Congenital Anomalies (EUROCAT)

Figure 1. Data collection overview

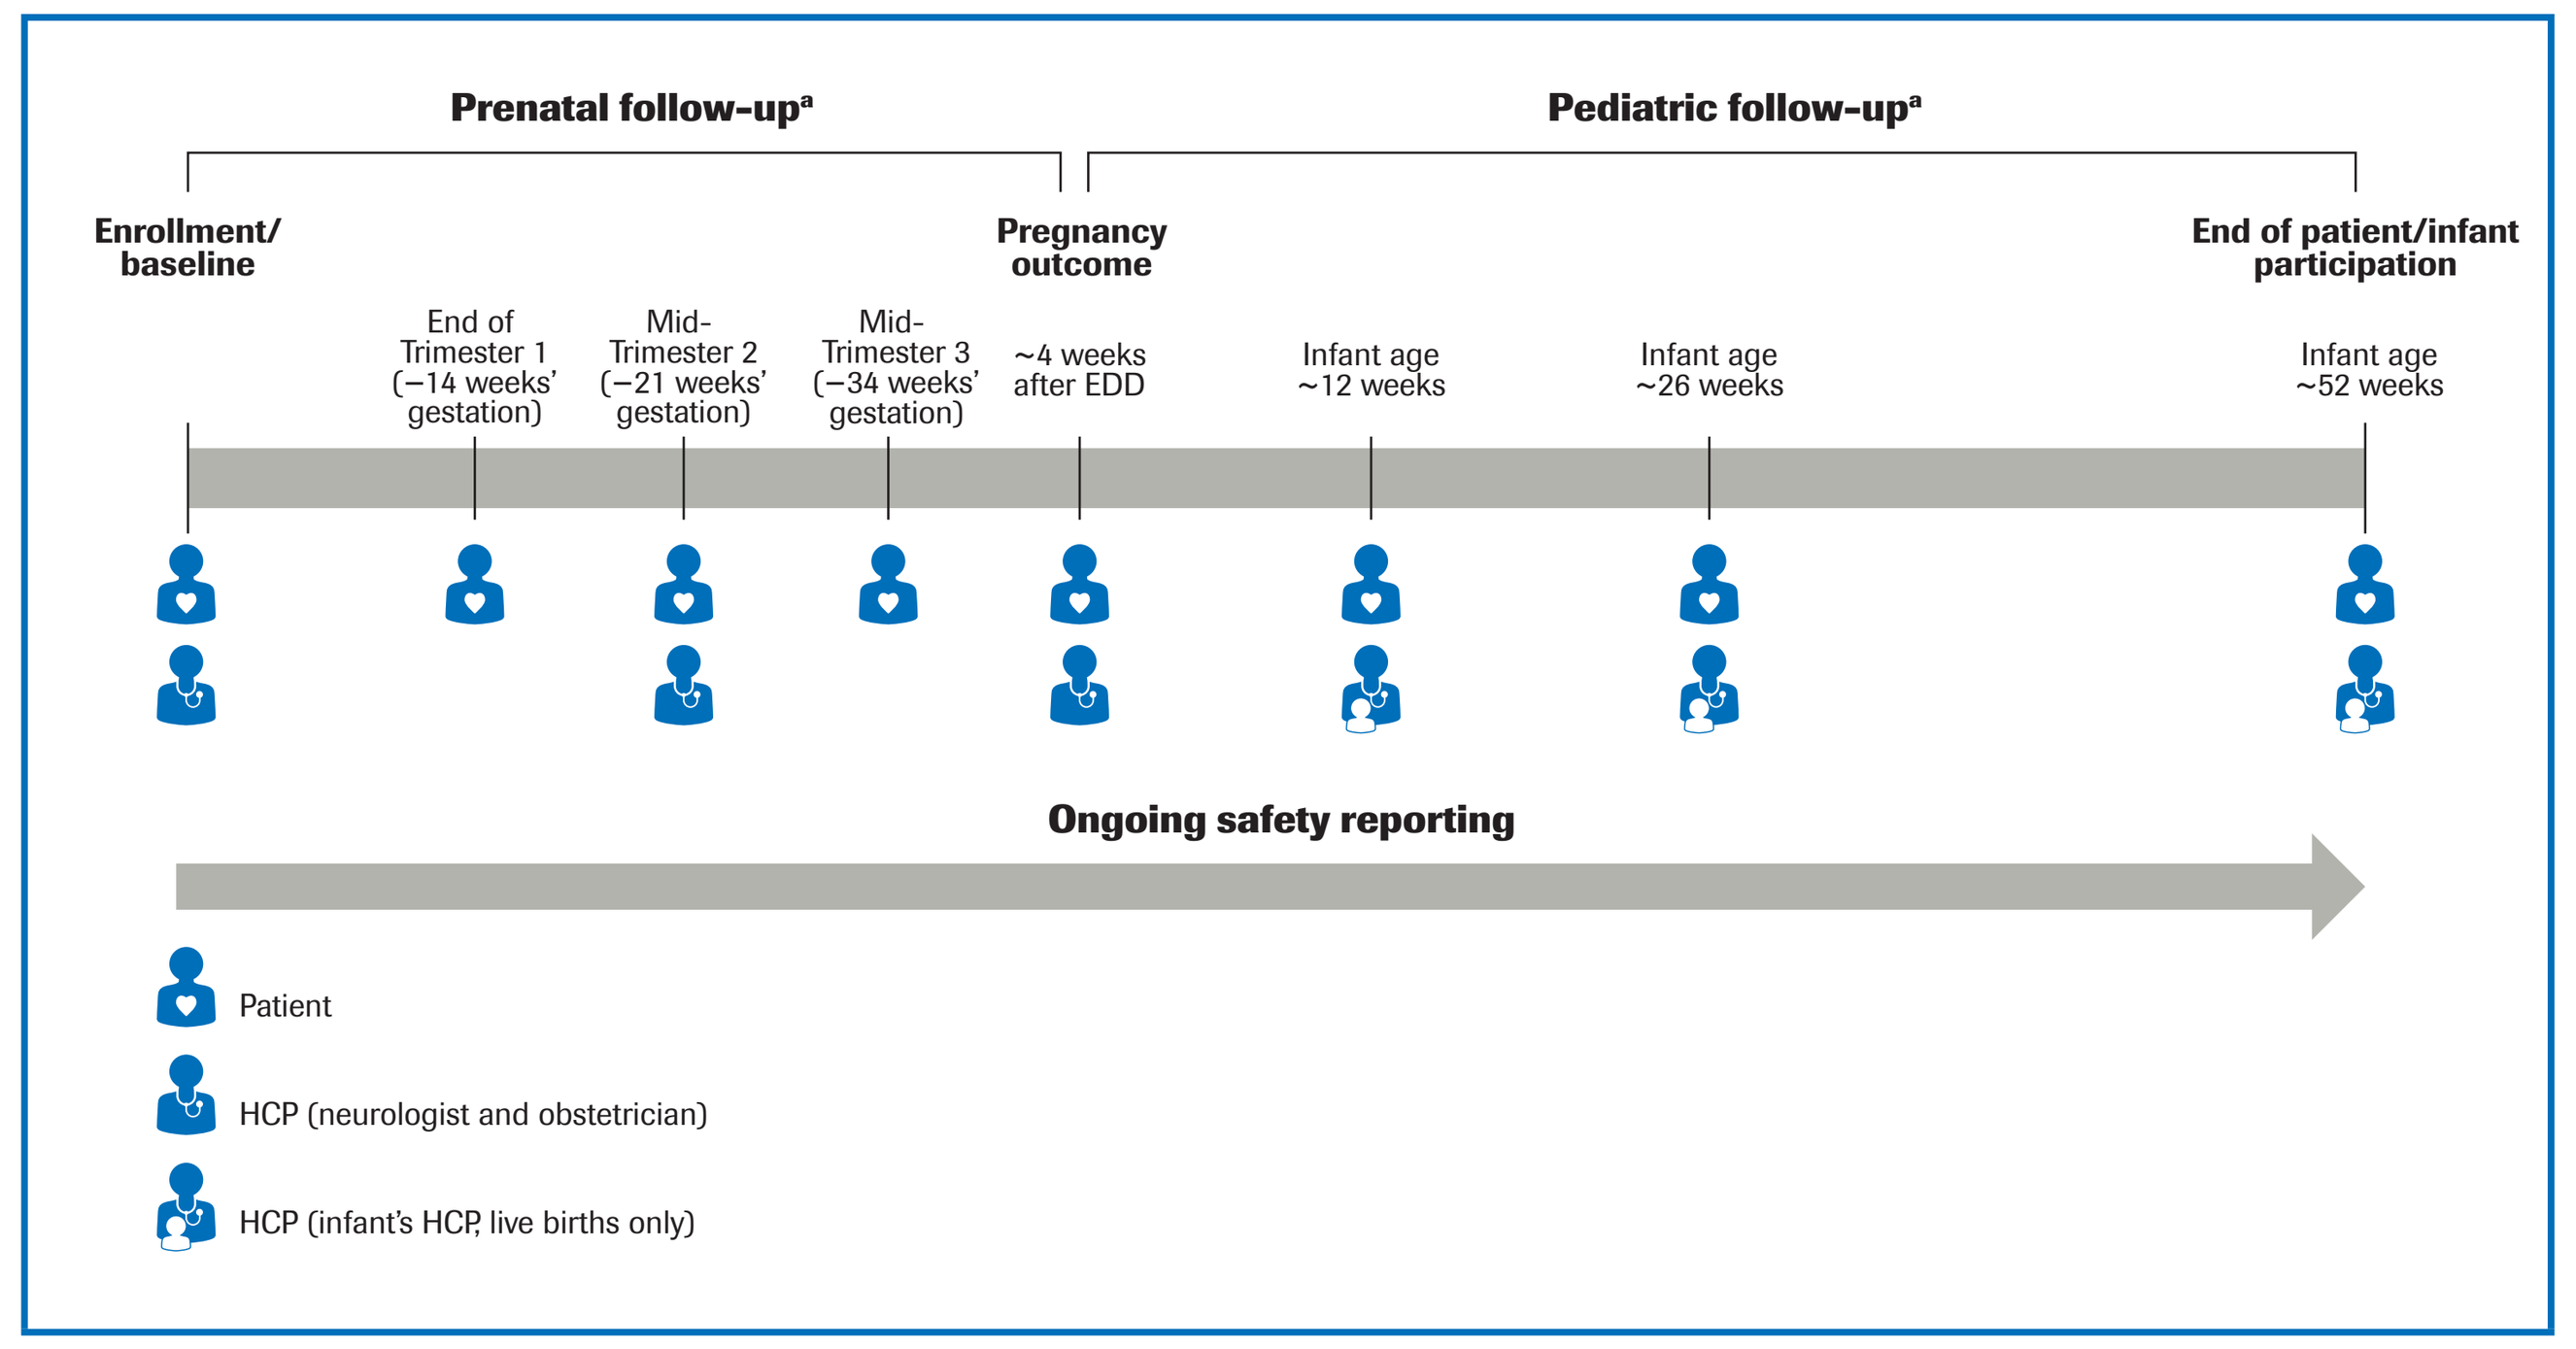

Prenatal follow-up[a]
Pediatric follow-up[a]
Enrollment/ baseline
End of Trimester 1 (~14 weeks' gestation)
Mid-Trimester 2 (~21 weeks' gestation)
Mid-Trimester 3 (~34 weeks' gestation)
Pregnancy outcome
~4 weeks after EDD
Infant age ~12 weeks
Infant age ~26 weeks
End of patient/infant participation
Infant age ~52 weeks
Ongoing safety reporting
Patient
HCP (neurologist and obstetrician)
HCP (infant's HCP, live births only)

[a]Timepoints of patient and HCP contact shown are based on expected intervals and may vary based on real-world observational data collection.
EDD, estimated delivery date; HCP, healthcare provider (e.g. neurologist, obstetrician, infant HCP, midwife).

Figure 2. Primary and secondary outcomes

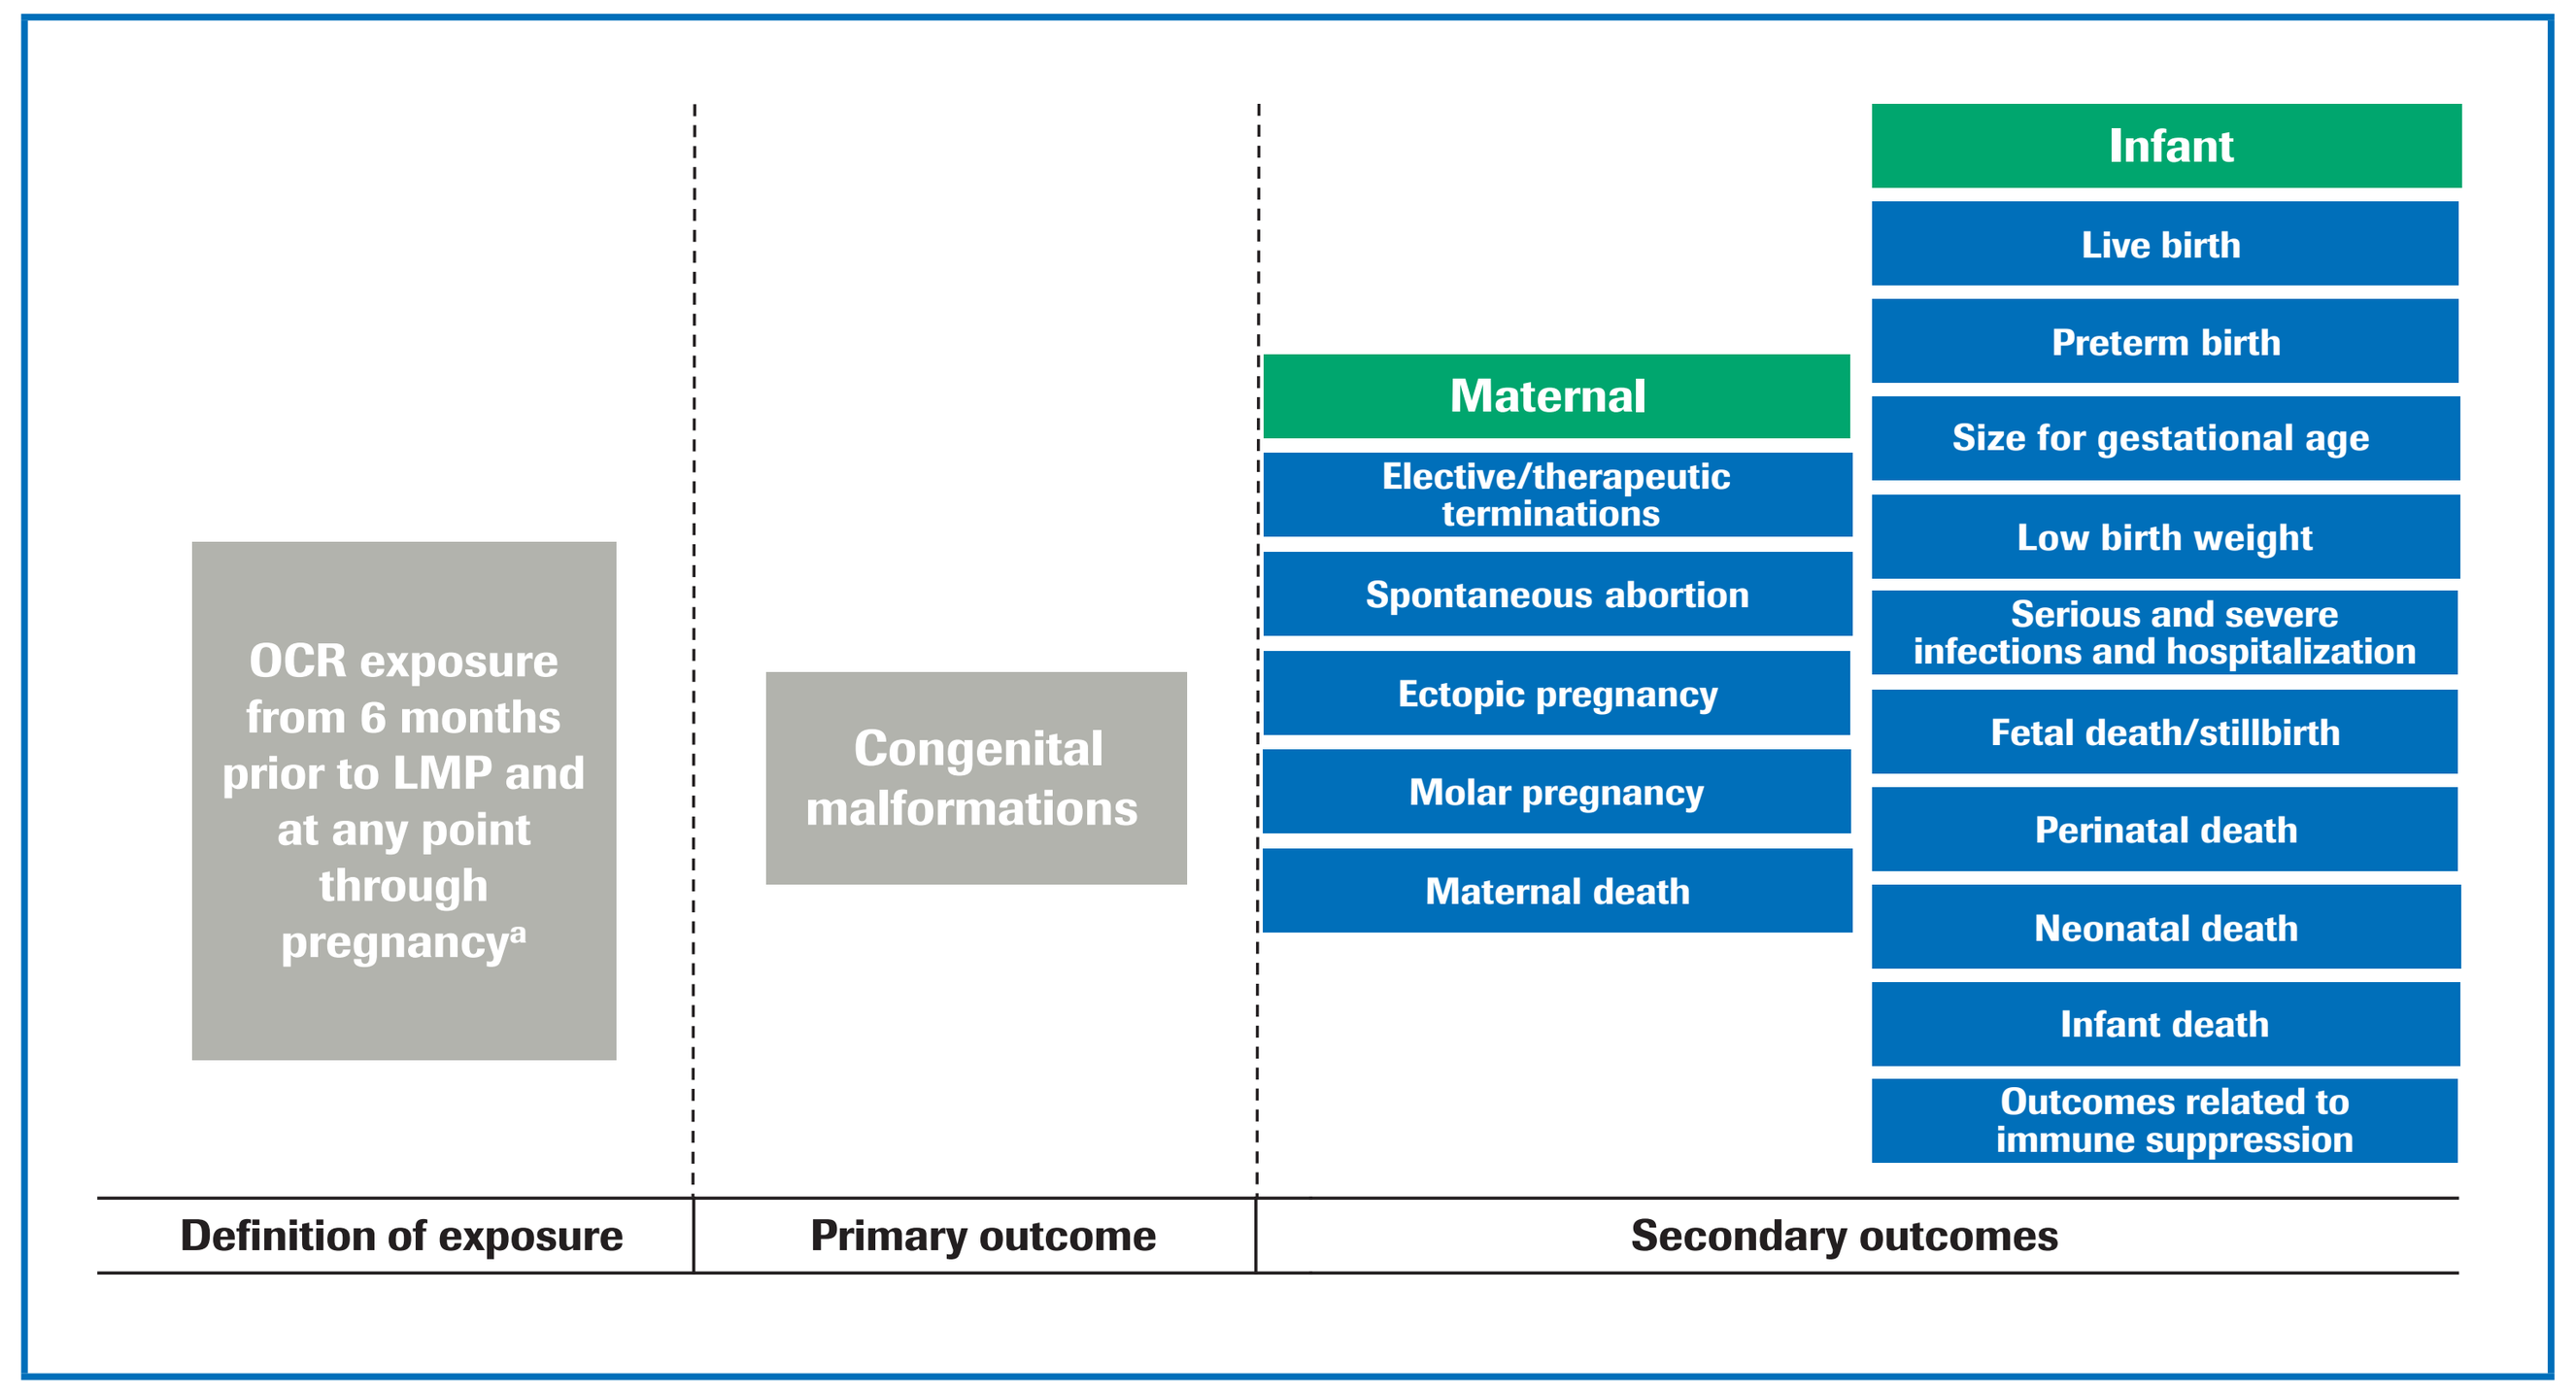

OCR exposure from 6 months prior to LMP and at any point through pregnancy[a]
Congenital malformations
Maternal
Elective/therapeutic terminations
Spontaneous abortion
Ectopic pregnancy
Molar pregnancy
Maternal death
Infant
Live birth
Preterm birth
Size for gestational age
Low birth weight
Serious and severe infections and hospitalization
Fetal death/stillbirth
Perinatal death
Neonatal death
Infant death
Outcomes related to immune suppression
Definition of exposure
Primary outcome
Secondary outcomes

[a]Including dosing and dates of administration during each trimester of pregnancy.
LMP, last menstrual period; OCR, ocrelizumab.

Eligibility Criteria

- Patients must meet the following criteria for study entry:
 - Currently pregnant
 - Diagnosed with MS
 - Documentation that the patient was exposed to OCR at any point starting from 6 months prior to LMP
 - The outcome of the pregnancy (e.g. pregnancy loss or live birth) must not be known

Table 1. Data collection

Baseline data collection	Follow-up during pregnancy
Patient demographics and characteristics	Changes in pregnancy status (gestational age, prenatal tests and results, etc.)
Current pregnancy information (LMP, gestational age, etc.)	Pregnancy outcome, if applicable
Maternal medical history ▪ Pregnancy history ▪ Surgical and medical history/significant maternal medical conditions other than MS ▪ MS disease history ▪ Family reproductive history ▪ Family MS history ▪ OCR treatment (start/stop dates, dosing, reasons for discontinuation, etc.) ▪ Current and prior medication use from 6 months prior to conception	Changes in MS disease status (including treatment changes, relapses, etc.) OCR prescription information Changes in comorbid conditions Current lifestyle factors (smoking, alcohol use, etc.) Current medications (including other MS treatments, medications with potential fetal health implications) SAEs related to pregnancy
Data collected at birth	**Pediatric follow-up**
Pregnancy outcome (live birth, stillbirth, elective termination, spontaneous abortion, etc.)	Feeding behavior (including breastfeeding)
Mode of birth (vaginal delivery, Cesarean section, etc.)	Weight, length
MS disease and treatment status since last follow-up	Developmental milestones
Changes in comorbid conditions since last follow-up	Laboratory values, if available
OCR prescription information	Evidence of any new congenital malformations since last follow-up
Current lifestyle factors (smoking, alcohol use, etc.)	Vaccination information
Current medications (including other MS treatments, medications with potential fetal health implications)	All infant SAEs (including serious and severe infections of NCI CTCAE severity Grade 3, 4, or 5 and hospitalizations other than for standard post-birth hospital stay)
Infant characteristics (gestational age at birth, sex, weight, height, congenital malformations noted, laboratory values [if available], etc.)	
SAEs related to pregnancy	
All infant SAEs (including serious and severe infections of NCI CTCAE severity Grade 3, 4, or 5 and hospitalizations other than for standard post-birth hospital stay)	

LMP, last menstrual period; MS, multiple sclerosis; NCI CTCAE, National Cancer Institute Common Terminology Criteria for Adverse Events; OCR, ocrelizumab; SAE, serious adverse event.

Sample Size

- Based on clinical, statistical and practical considerations, 92 pregnancy outcomes are required to achieve a minimum 80% power at a significance level of α=0.05 to detect a relative risk of 3 in major congenital malformations, major birth defects and preterm births relative to the baseline prevalence (**Table 2**)
- A live birth rate of 91% was observed in MS pregnancy registries in Italy and Germany;[9,10] a rate of ~62% was observed in a general population, not restricted to MS[5,11]
- Assuming a 15% drop-out rate , at least 120, 142 or 176 pregnancies would need to be enrolled to have 92 live birth pregnancy outcomes, depending on the proportion of pregnancies resulting in live births (91%, 77% or 62%, respectively)

Table 2. Power to detect the prevalence of adverse pregnancy outcomes

	Power/adverse pregnancy prevalence		
Relative risk	3% baseline prevalence (MCMs)[12]	4% baseline prevalence (major birth defects in OCR-exposed patients)[13]	10% baseline prevalence (preterm births)[14]
2.5	64.59%	73.90%	97.53%
3.0	80.10%	88.11%	99.81%
3.5	89.57%	95.13%	99.99%

Note: power is calculated using 2-sided exact test at α=0.05 significance level for 1 proportion.
Sample size and power calculations are performed using PASS software version 14.
MCM, major congenital malformation; OCR, ocrelizumab.

RESULTS

- The planned start date is mid-2018 and key study milestones are shown in **Figure 3**
- The total duration of participation is 21 months, and the study will last approximately 10 years
- Interim results will be communicated when sufficient patients have been accrued to allow meaningful analysis and final results will also be communicated to the MS community

Figure 3. Study milestones

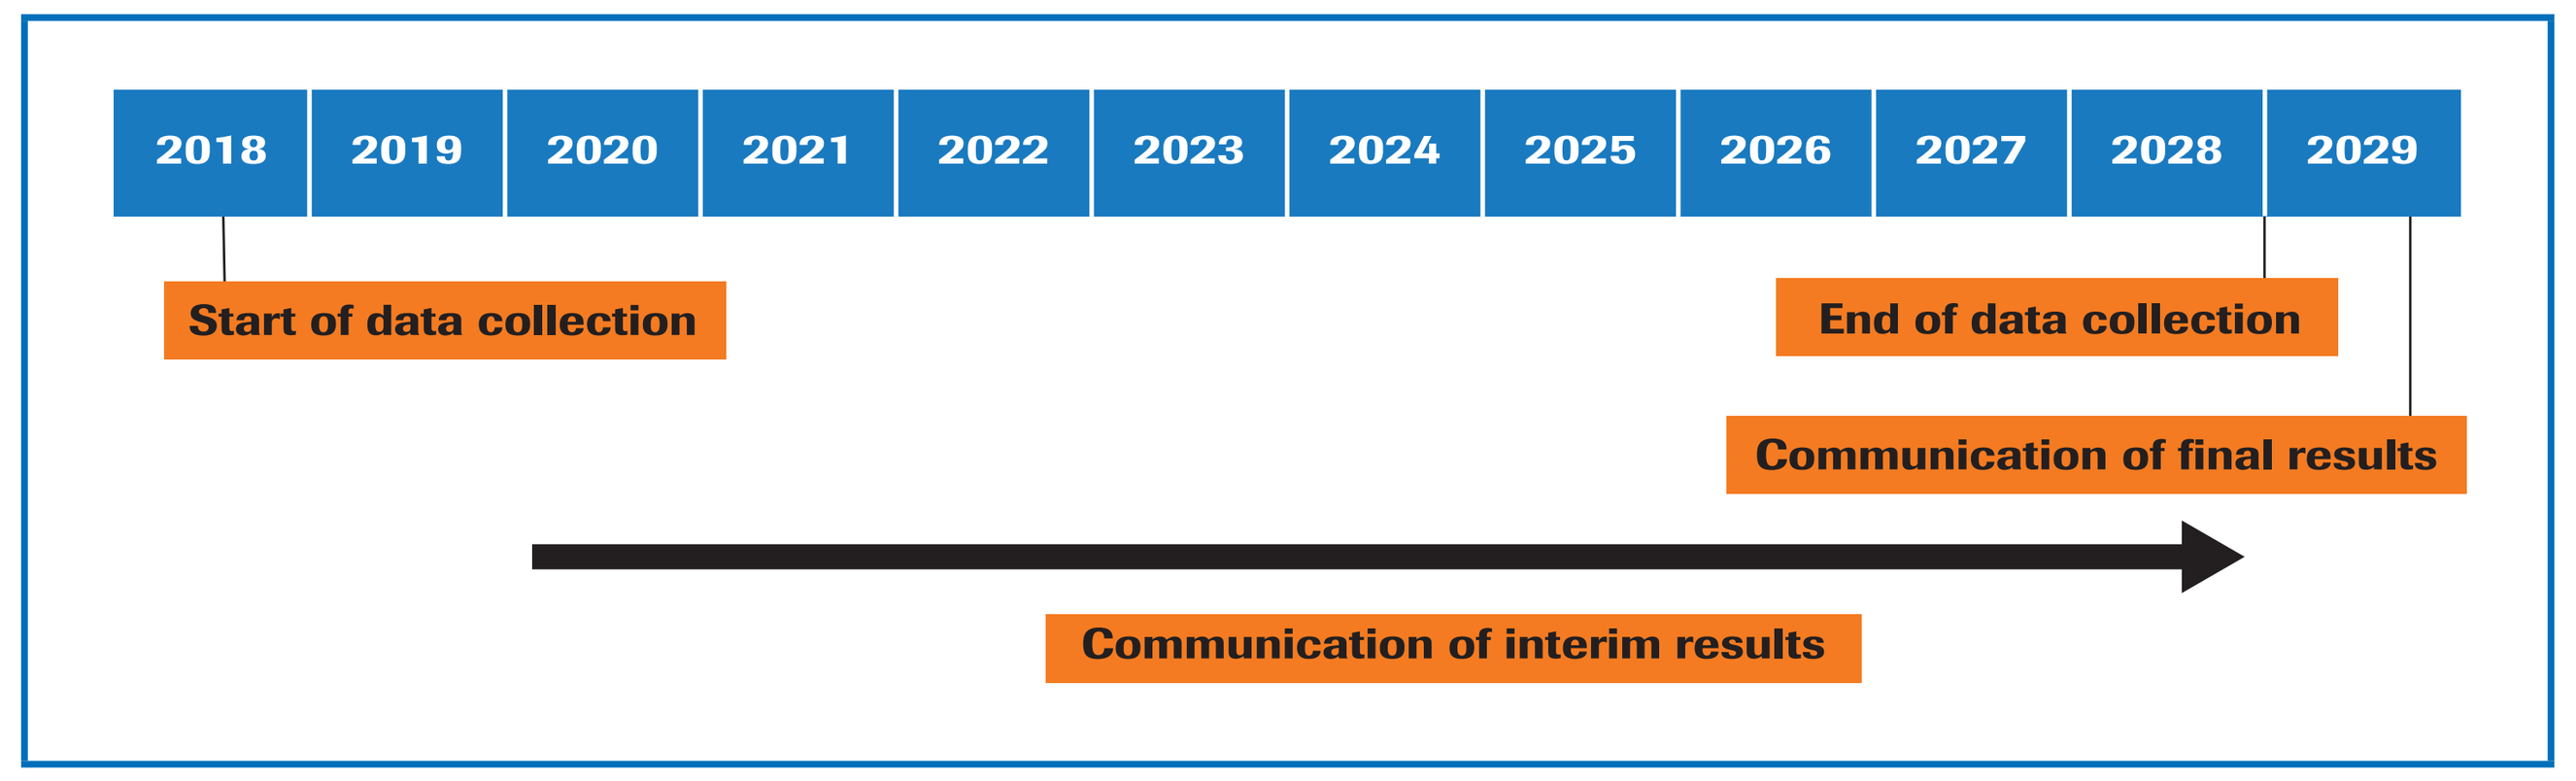

2018
2019
2020
2021
2022
2023
2024
2025
2026
2027
2028
2029
Start of data collection
End of data collection
Communication of final results
Communication of interim results

CONCLUSIONS

- The Ocrelizumab Pregnancy Registry is a multicenter, prospective, observational study that will provide insights on the safety profile of ocrelizumab during pregnancy in a real-world setting and complement the multi-source post-marketing study (see Poster 372[15]) by providing detailed case information early after approval
- This information is important for patients, clinicians and healthcare decision-makers and will support discussions with pregnant women with MS who are planning to become pregnant or may have been exposed to ocrelizumab before or during pregnancy

ACKNOWLEDGMENTS

This research is being funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland. Writing and editorial assistance for this presentation was provided by Articulate Science, UK, and funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

REFERENCES

1. Kane SV, Acquah LA. *Am J Gastroenterol* 2009;104:228–233.
2. Vukusic S, *et al. ECTRIMS/ACTRIMS* 2017;Poster presentation P710.
3. EMEA. Ocrevus (ocrelizumab). Summary of Product Characteristics. http://www.ema.europa.eu/ema/index.jsp?curl=pages/medicines/human/medicines/004043/human_med_002187.jsp&mid=WC0b01ac058001d124.
4. FDA. OCREVUS™ (ocrelizumab). Prescribing Information. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2017/761053Orig1s000Lbl.pdf.
5. FDA. Establishing Pregnancy Exposure Registries. https://www.fda.gov/downloads/drugs/guidancecomplianceregulatoryinformation/guidances/ucm071639.pdf.
6. EMEA. Guideline on the exposure to medicinal products during pregnancy: need for post-authorisation data. http://www.ema.europa.eu/docs/en_GB/document_library/Regulatory_and_procedural_guideline/2009/11/WC500011303.pdf.
7. Gliklich R, *et al.* Registries for Evaluating Patient Outcomes: A User's Guide. https://effectivehealthcare.ahrq.gov/sites/default/files/pdf/registries-guide-3rd-edition_research.pdf.
8. ISPE. *Pharmacoepidemiol Drug Saf* 2008;17:200–208.
9. Herbstritt S, *et al. Mult Scler* 2016;22:810–816.
10. Thiel S, *et al. Mult Scler* 2016;22:801–809.
11. Ventura SJ, *et al. Vital Health Stat 21* 2000;(56):1–47.
12. CDC. MMWR Morb Mortal Wkly Rep. 2008;57:1–5.
13. Roche. Data on file.
14. Ferré C, *et al. MMWR Morb Mortal Wkly Rep* 2016;65:1181–1184.
15. Margulis AV, *et al. AAN* 2018;Abstract P4.372.

DISCLOSURES

D Wormser is an employee and shareholder of F. Hoffmann-La Roche Ltd. P Engel is an employee of IQVIA. K Hahn is an employee of QuintilesIMS. S Bader-Weder is an employee and shareholder of F. Hoffmann-La Roche Ltd. E-M Didden is an employee of F. Hoffmann-La Roche Ltd. J Evershed is an employee of Roche Products Ltd and shareholder of F. Hoffmann-La Roche Ltd. M Garas is an employee and shareholder of F. Hoffmann-La Roche Ltd. Q Wang is an employee of F. Hoffmann-La Roche Ltd. K Hellwig reports funding from Roche, Novartis, Teva, Bayer, Merck, Genzyme and Biogen.

OCR/032218/0083e

Presented at the 70th American Academy of Neurology (AAN) Annual Meeting; April 21–27, 2018; Los Angeles, CA, USA

Poster 367